EUROPEAN HEMATOLOGY ASSOCIATION

EHA2024 JUNE 13 - 16 | MADRID

INTEGRATION OF POINT-OF-CARE TESTING IN PRIMARY HEALTH CARE FACILITIES FOR EARLY COMPREHENSIVE MANAGEMENT OF SICKLE CELL DISEASE IN RURAL KASSENA NANKANA DISTRICTS OF GHANA.

E. DUAH[1], L. OSEI ABABIO[1], S.T CHATIO[1], L. NICOLA[2], O. EGBUJO[5], K. MARFO[2], W. DORMECHELE[1], P. ANSAH[1] and I. ODAME[3,4]

1Navrongo Health Research Center, upper East, Ghana 2Global Health & Sustainability, Novartis Pharma AG, Basel, Switzerland

3The Hospital for Sick Children, Toronto, ON, Canada 4Department of Pediatrics, University of Toronto, Toronto, ON. Canada

5Novartis Pharmaceuticals Corporation, New Jersey, USA

INTRODUCTION

- Sickle cell disease (SCD) mortality rates are highest in children under- 5. WHO estimates that SCD contributes to 9-16% of under-5 mortality in certain regions of West Africa, imposing physiological, mental, and financial burdens on affected individuals and families.
- Mortality and morbidity can be reduced through early diagnosis and comprehensive care.
- In sub-Saharan Africa, limited access to specialized care facilities and the high cost of laboratory-based diagnostic tests have hindered early diagnosis and timely interventions for children with SCD.
- Point-of-care testing (POCT) devices may be advantageous because of immediate screening results at testing sites, ease of use in remote areas and immediate linkage into comprehensive care.

AIM

Our study evaluated the feasibility and acceptability of integrating point-of-care testing for sickle cell disease at primary healthcare facilities and the effectiveness of linking screen-positive children to early comprehensive SCD care.

METHOD

- This prospective observational study utilized implementation science methods to integrate the POCT device (Gazelle), based on a micro-engineered hemoglobin (Hb) electrophoresis technique, as a screening tool for SCD in primary healthcare settings.
- Confirmatory testing was performed at the first clinic visit using a lateral flow immunoassay POCT device (Sickle SCAN).
- The study was conducted in health facilities in the Kassena Nankana Municipal and Kassena Nankana West Districts of the Upper East Region. These included four Community Health and Planning Services (CHPS) compounds, five health centers, and one district hospital. All children under- 5 years were screened after obtaining written consent from their caregivers.
- All children identified with SCD were referred to the SCD clinic at the district hospital for comprehensive care. IRB and Ethics approval was obtained.

RESULTS

- Between August 2022 and September 2023, 6024 consecutive newborns and infants under- 5 were screened.
- Our findings show that out of 6024 children, 3971 (66%) had normal hemoglobin (HbAA), 1146 (19%) had HbAC, 720 (11.9%) had HbAS, 106 (1.8%) had HbCC, **58 (1.0%) had HbSC,** and **23 (0.3%) had HbSS.**
- All 81 children who tested positive on the screening completed the confirmatory diagnosis during their first visit to the SCD clinic.
- Except for two children who relocated to another region, all 79 participants were retained in care with no dropouts, facilitated by periodic home visits from community health nurses at the various testing sites.
- Thus, 81 (1.3%) children had SCD, 79 of whom received penicillin V prophylaxis and folic acid as part of their care. 23 HbSS patients with a mean age of 23.4 ± 17.1 months were initiated on hydroxyurea therapy. 43% of the babies had their hydroxyurea initiated before their first birthday.
- All patients continue with follow-up care facilitated by community health workers and Vula Mobile (telehealth referral platform) for long-term clinical outcomes.

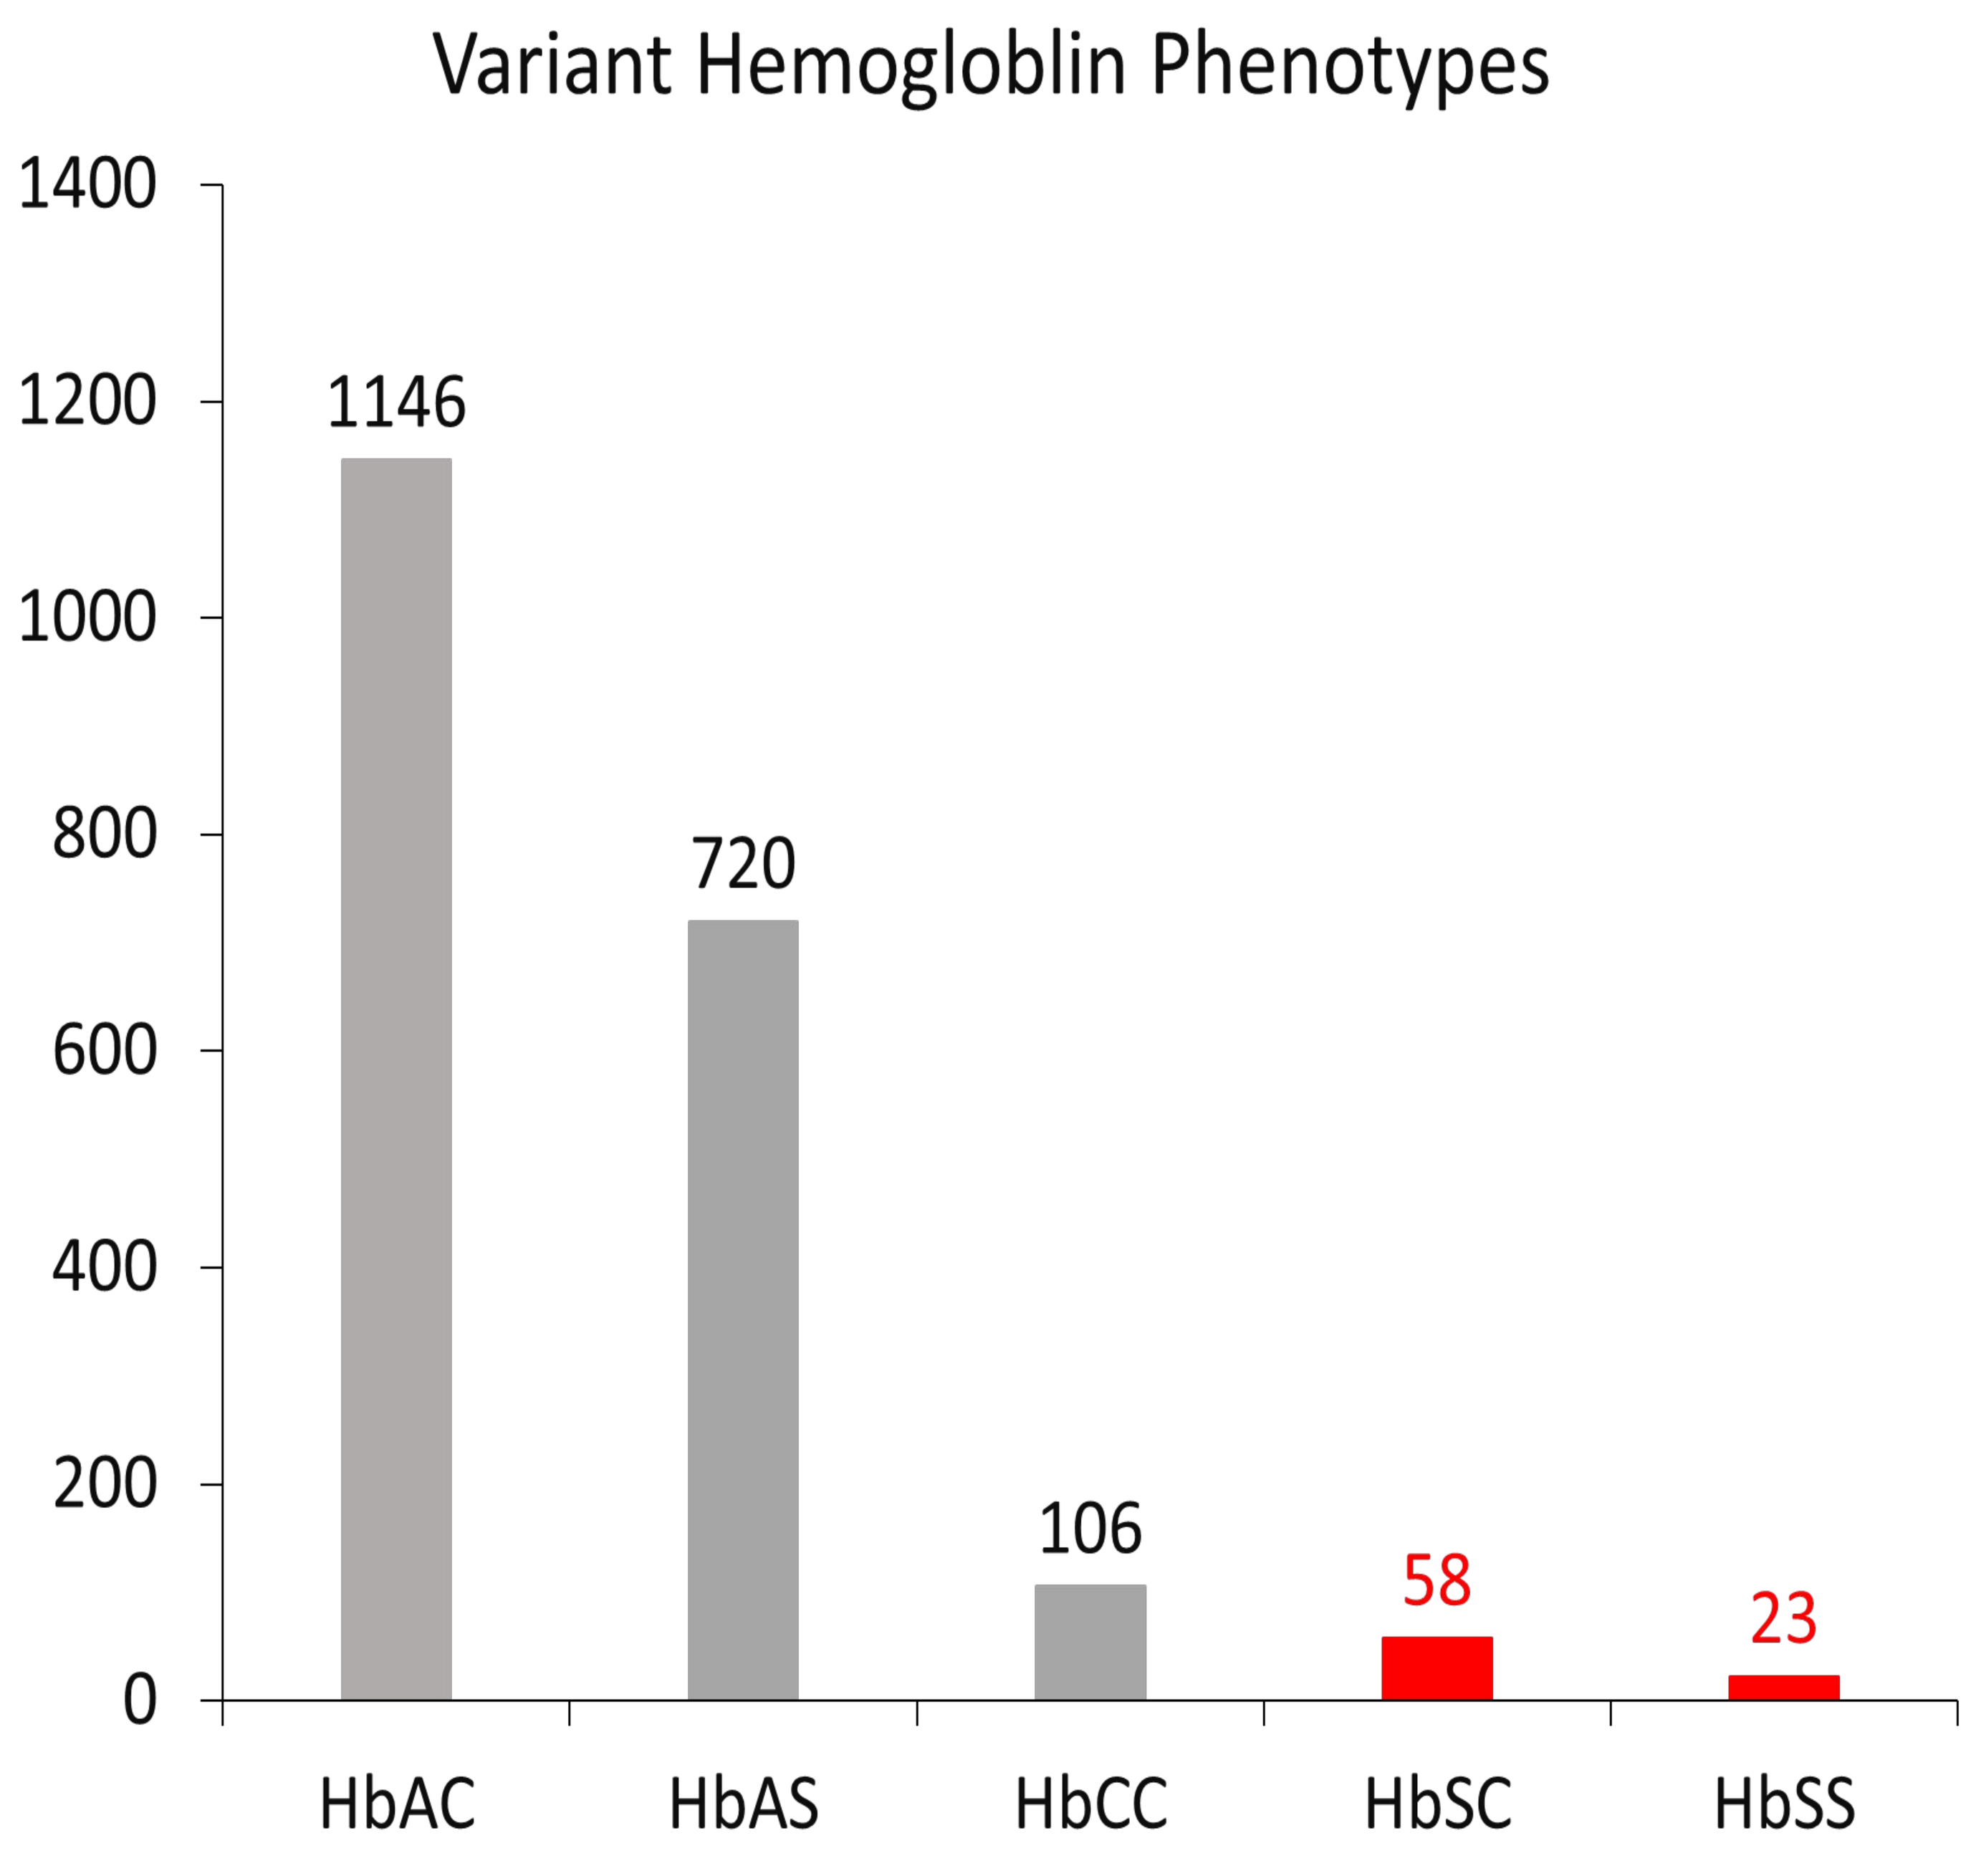

Variant Hemoglobin Phenotypes
1400
1200
1000
800
600
400
200
0
1146
720
106
58
23
HbAC
HbAS
HbCC
HbSC
HbSS

Figure 1: Variant Hemoglobin phenotypes in the children screened for SCD.

Variable	Frequency (N=6024)	Percentage (%)
Age category (months)		
0 – 11	2483	41.2
12 – 23	1377	22.9
24 – 35	885	14.7
36 – 47	734	12.2
48 – 59	545	9.0
Gender		
Female	2992	49.7
Male	3032	50.3

Table 1: Number and proportions of children screened for SCD by age group and gender.

CONCLUSIONS

- Our findings demonstrated that integrating POCT screening into existing primary healthcare programs, leveraging the high immunization coverage, is feasible and can be rapidly implemented in remote areas with limited resources.
- The prevalence rate from this study is comparable to previous screening efforts with laboratory-based techniques in other parts of the country.
- All screen-positive patients, except for two who relocated to another region, were referred for SCD management, and adherence to the first clinic visit was 100%.
- We continue to provide follow-up care to the patients and monitor long-term clinical outcomes. The insights gained from this study will be utilized to expand to other areas of the district and the entire Upper East Region of Ghana.

REFERENCES

1. Nnodu O-E et al. Implementing newborn screening for sickle cell disease as part of immunisation programmes in Nigeria: a feasibility study. Lancet Haematol. 2020;7(7):e534–40.
2. Thomson AM, McHugh TA, Oron AP, Teply C, Lonberg N, Vilchis Tella V, et al. Global, regional, and national prevalence and mortality burden of sickle cell disease, 2000–2021: a systematic analysis from the Global Burden of Disease Study 2021. Lancet Haematol. 2023;10(8):e585–99.
3. Grosse SD, Odame I, Atrash HK, Amendah DD, Piel FB, Williams TN. Sickle cell disease in Africa: A neglected cause of early childhood mortality. Vol. 41, American Journal of Preventive Medicine. 2011.
4. Newborn Screening for Sickle Cell Disease in Liberia A Pilot Study - Tubman - 2016 - Pediatric Blood & Cancer - Wiley Online Library.
5. Odame I. We need a global solution. Nature. 2014;515(7526):S10.

ACKNOWLEDGEMENT

- The authors acknowledge the hard work of all the nurses and midwives in the facilities involved in this study.
- We are deeply grateful to the dedicated staff of the sickle cell clinic at War Memorial Hospital, Navrongo.
- We are grateful to all the participants for their valuable contributions to this study.
- This study was part of a research collaboration, with funding provided by Novartis Pharma AG to Hospital for Sick Children and Navrongo Health Research Center.

CONTACT INFORMATION

Lucy Osei Ababio (MBChB)

Telephone: +233 207804309

Email: lucyoseiababio@gmail.com